Evaluation of immune checkpoint inhibitors and targeted therapies ex vivo in live tumor fragments from core biopsies and resections accurately predicts response in the clinic

Sean Caenepeel,[1] Pichet Adstamongkonkul,[1] Yuan-I Nina Chen,[1] Pierre Cunin,[1] Amreen Nasreen,[1] Christin Johnson,[1] MaryAnne Evans,[1] Brett Duffin,[1] Nathan Marhefke,[1] Chetan Sood,[1] Nicholas Dana,[1] Aishwarya Sunil,[1] Laura Hrycyniak,[1] Julie Zweng,[1] Erika von Euw,[1] Christina Vivelo,[1] Hinco Gierman,[1] Hilario Ramos,[1] Premal Patel[1]

[1]Elephas Biosciences, Madison, WI

Introduction

- Current biomarkers for predicting which patients will benefit from immune checkpoint inhibitors (ICIs) and KRAS-targeted therapies are imperfect[1,2]
- Ex vivo profiling of live tumor tissue accurately models the tumor microenvironment (TME) and enables response prediction within days[3,4]
- The elive platform profiles live tumor tissue and preserves the TME through 72 hours, enabling rapid characterization of responses to ICIs and targeted therapies

Conclusions

- The elive platform accurately predicts outcomes to ICIs by cytokine profiling and is therefore positioned to characterize responses to immunotherapies with diverse mechanisms of action
- Development of a cytotoxicity endpoint expands the elive platform's potential applications beyond ICIs to include the evaluation of additional therapeutics, such as targeted therapies

Acknowledgments

- We thank the patients, clinicians, research staff, and Elephas employees for their contributions
- Tissue samples were provided by the Cooperative Human Tissue Network, which is funded by the National Cancer Institute

Methods

1 Functional profiling of live tumor fragments on the elive platform

A

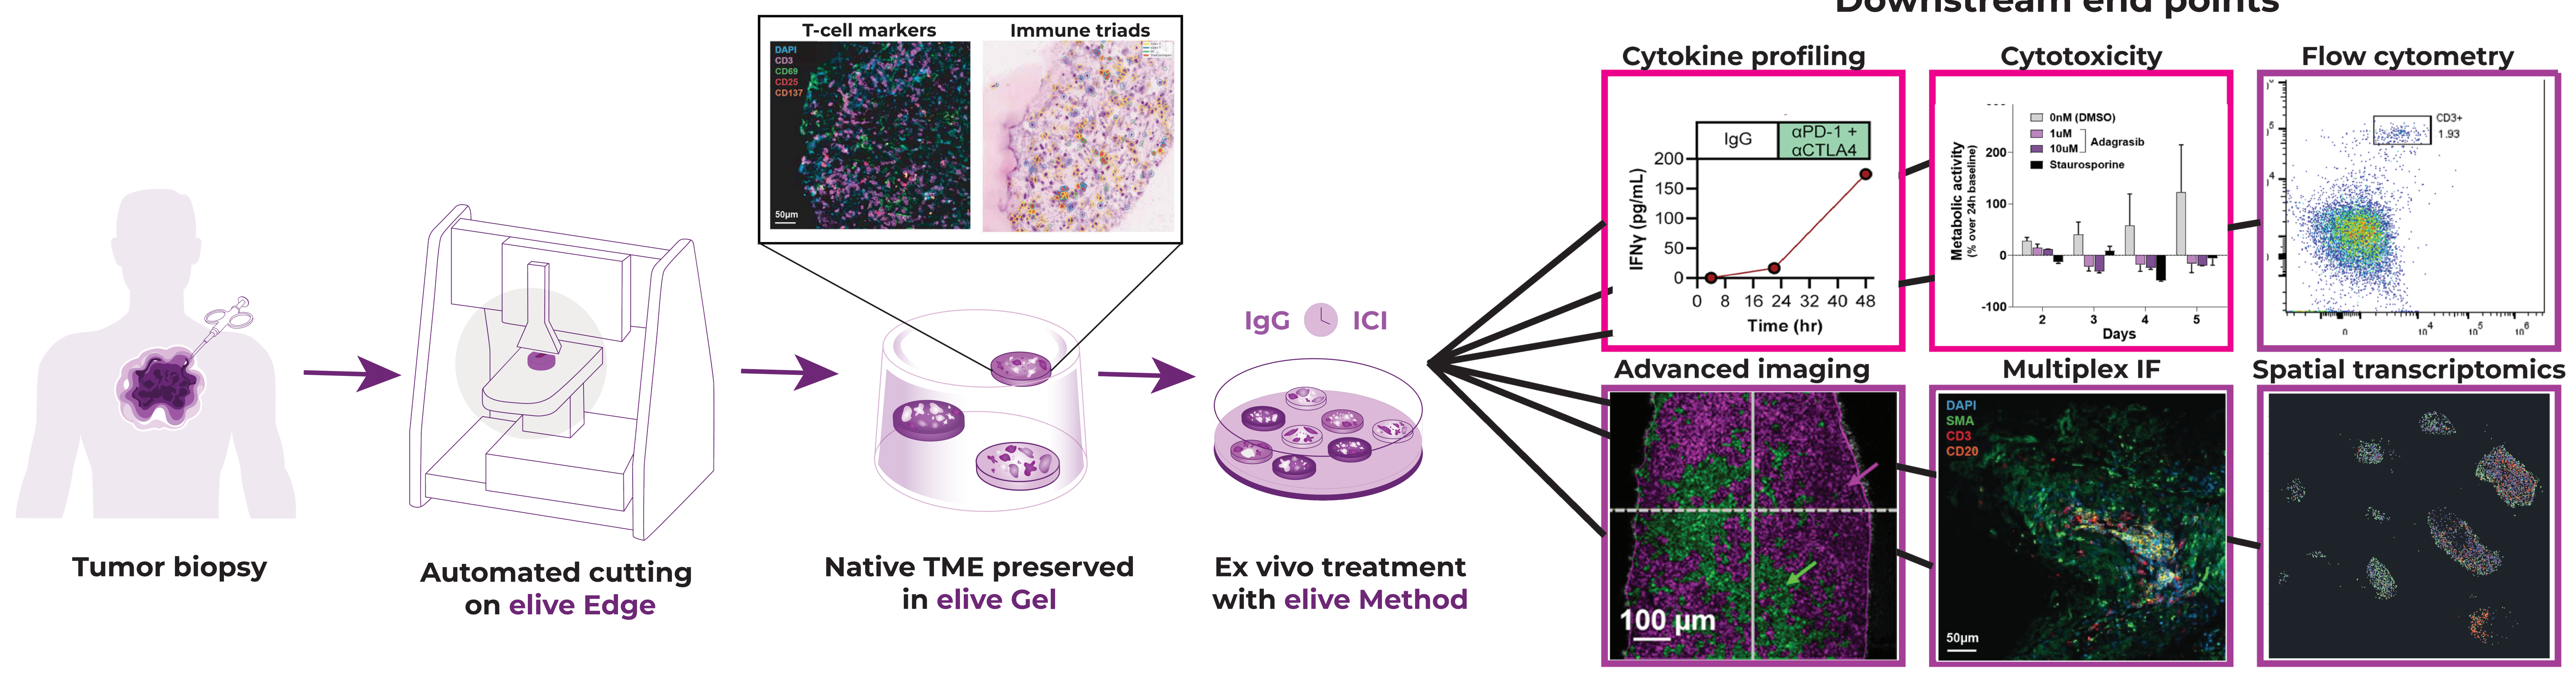
Tumor biopsy
Automated cutting on elive Edge
T-cell markers
Immune triads
DAPI
CD3
CD69
CD25
CD137
50µm
Native TME preserved in elive Gel
IgG
ICI
Ex vivo treatment with elive Method
Downstream end points
Cytokine profiling
IgG
αPD-1 + αCTLA4
IFNγ (pg/mL)
Time (hr)
Cytotoxicity
0nM (DMSO)
1uM
10uM
Adagrasib
Staurosporine
Metabolic activity (% over 24h baseline)
Days
Flow cytometry
CD3+
1.93
Advanced imaging
100 µm
Multiplex IF
DAPI
SMA
CD3
CD20
50µm
Spatial transcriptomics

B

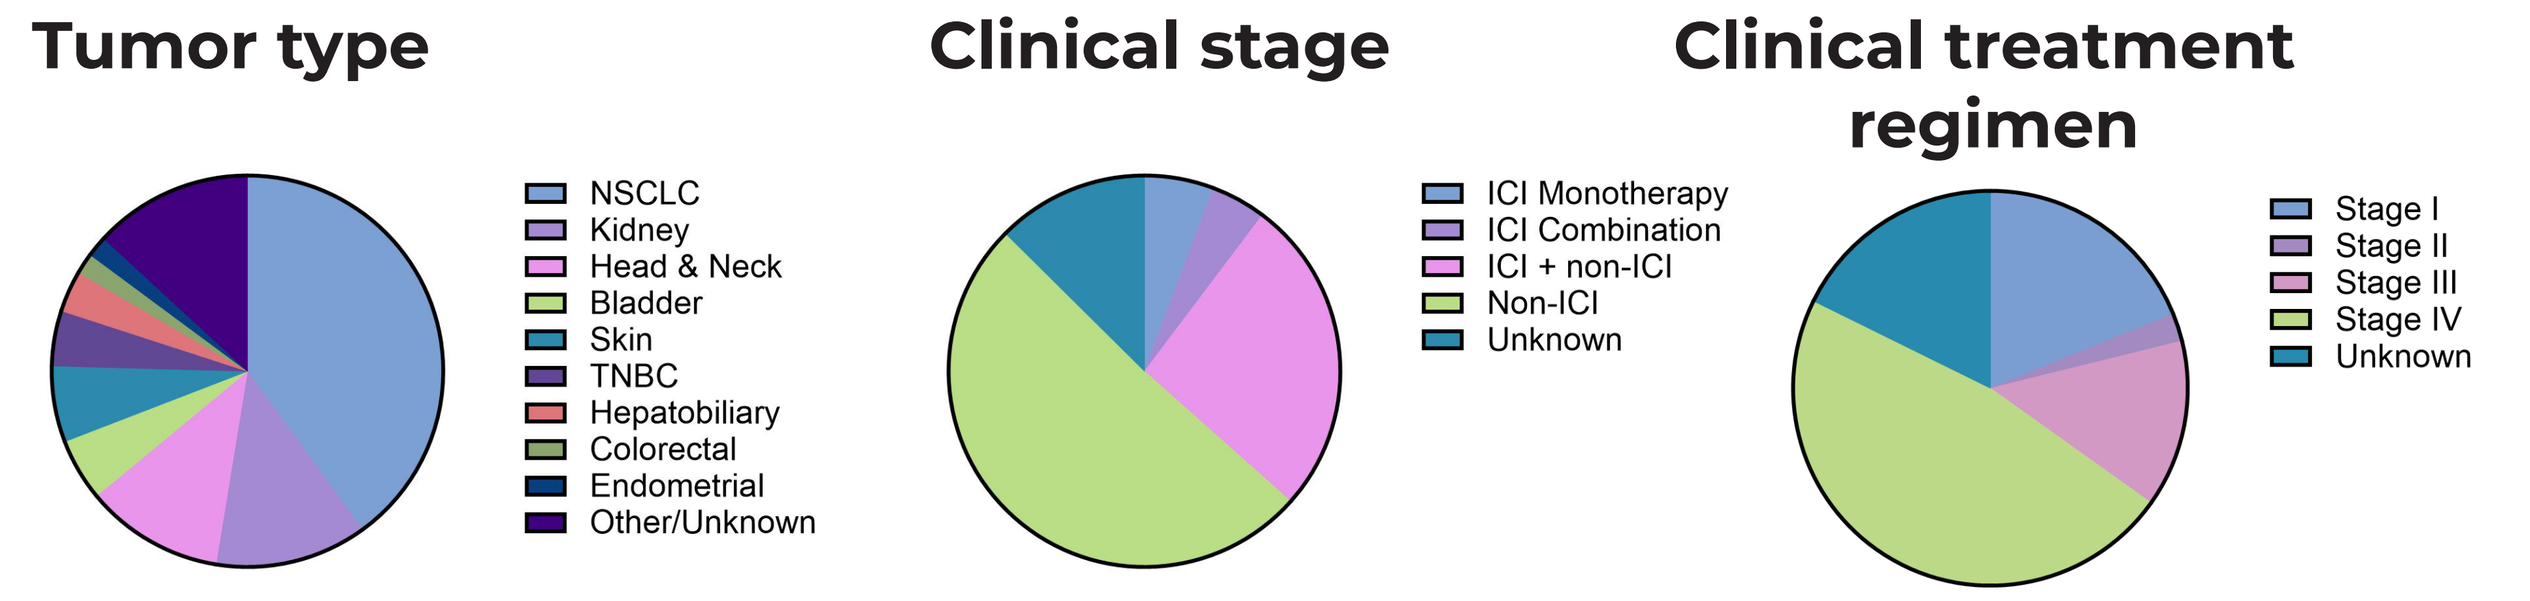
Tumor type
NSCLC
Kidney
Head & Neck
Bladder
Skin
TNBC
Hepatobiliary
Colorectal
Endometrial
Other/Unknown
Clinical stage
ICI Monotherapy
ICI Combination
ICI + non-ICI
Non-ICI
Unknown
Clinical treatment regimen
Stage I
Stage II
Stage III
Stage IV
Unknown

C

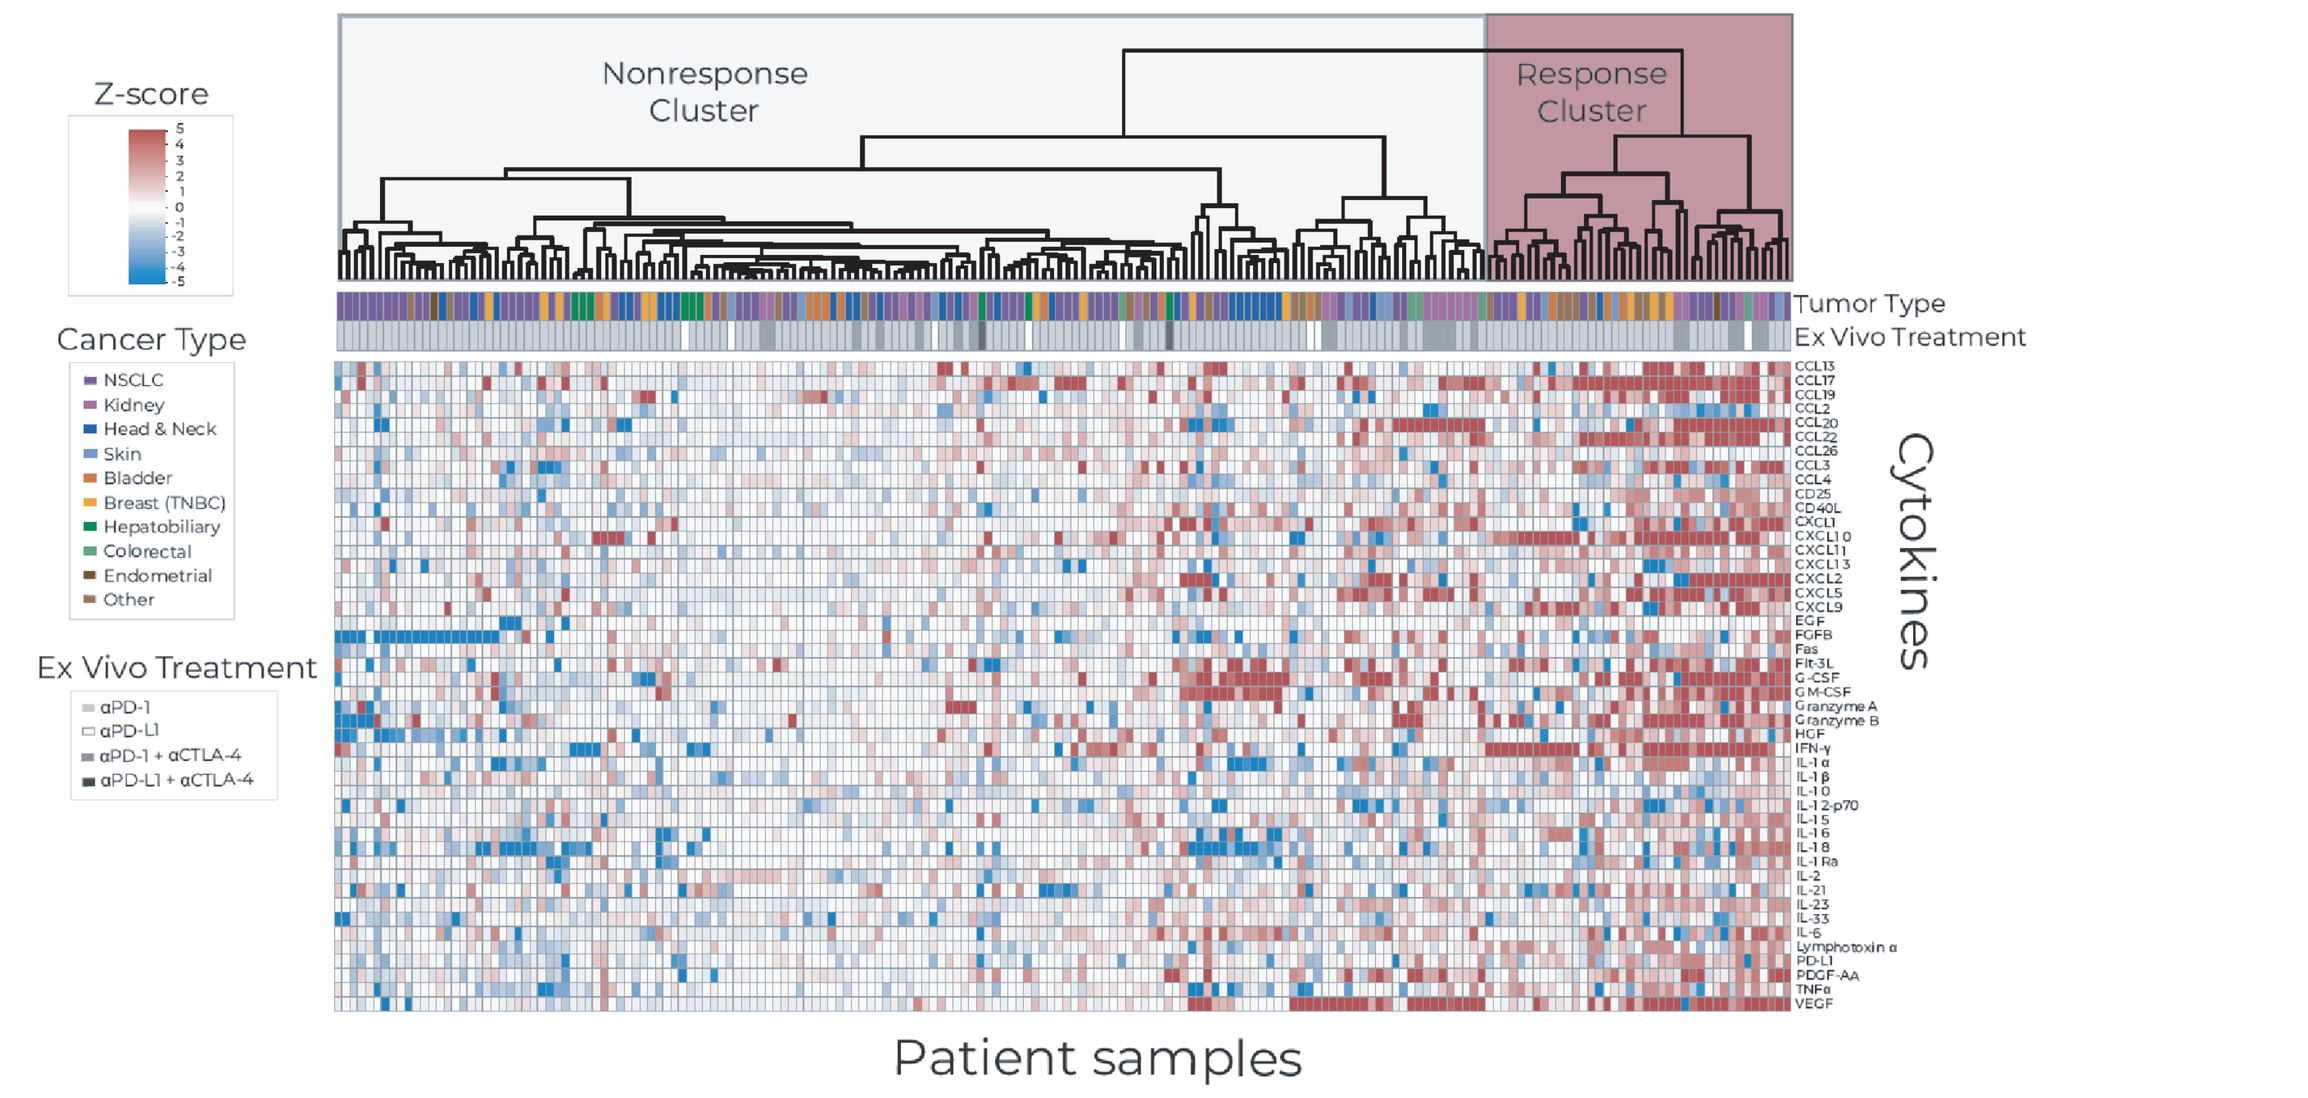
Z-score
Cancer Type
Ex Vivo Treatment
Nonresponse Cluster
Response Cluster
Tumor Type
Ex Vivo Treatment
Cytokines
Patient samples

A. The elive platform is used for functional profiling of live tumor fragments (LTFs) treated ex vivo with ICIs or cytotoxic agents. Fresh tumor tissue from standard of care biopsies is processed within 24 hours of collection. Biopsy tissue is cut into LTFs using elive Edge, a bespoke automated cutting instrument, and encapsulated in elive Gel, a proprietary hydrogel that preserves the native TME. Once encapsulated, LTFs are treated ex vivo using a sequential treatment strategy in which IgG control followed by ICI is added to the same tissue in a single well to mitigate variability from tumor heterogeneity. Longitudinal multiplex cytokine profiling is performed to assess tumor response to ex vivo ICI treatment. The elive platform is compatible with a wide range of downstream end points, including cytotoxicity assays, flow cytometry, advanced imaging, multiplex immunofluorescence, and spatial transcriptomics. **B.** To interrogate cytokine profiling as a measure of ex vivo response to ICI, tumor tissue and clinical data were collected from patients with solid tumors who were eligible for ICI treatment and enrolled in Elephas clinical trials (NCT05478538, NCT05520099, NCT06349642, NCT07327489; N = 175 patients).[5] Tumors collected were primarily non small cell lung cancer (NSCLC), kidney, and head and neck.[5] Most patients had stage IV disease, and nearly one third of patients went on to receive some form of ICI treatment.[5] **C.** Unsupervised hierarchical clustering of cytokine profiling data from LTFs (n = 186 samples from 124 tumors) revealed cytokine response and nonresponse clusters. The heatmap shows the delta of the change in cytokine production rates between the ICI and IgG treatment phases (z score normalized), which were used to develop a predictive algorithm.[5]

Results

2 The elive platform accurately predicts clinical response to ICIs[5]

A

Analyte	Normalized threshold	Weight	HM sensitivity	HM specificity
IFN-γ	4.7	2	0.74	0.97
CCL22	3.2	2	0.54	0.97
CXCL10	2.8	2	0.74	0.93
Lymphotoxin-α	1.5	2	0.51	0.99
TNF-α	0.9	2	0.59	0.93
CCL17	8.4	1	0.46	0.97
Granzyme B	1.2	1	0.77	0.83
IL-10	0.4	1	0.72	0.83

B

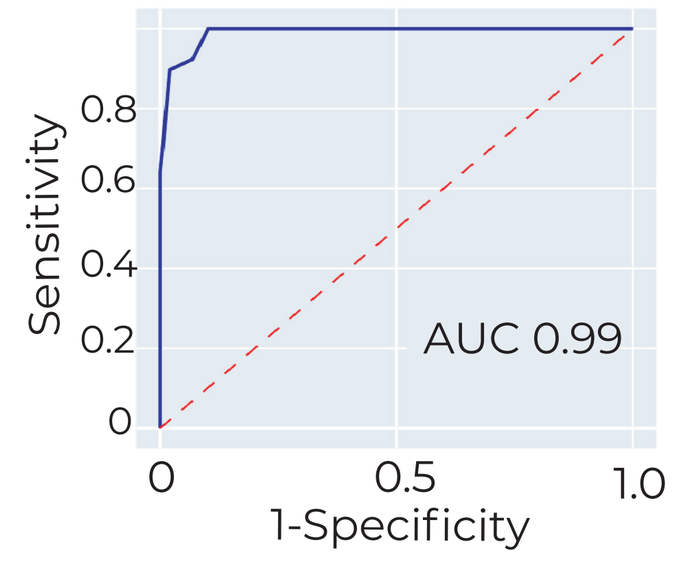
Sensitivity
1-Specificity
AUC 0.99

C

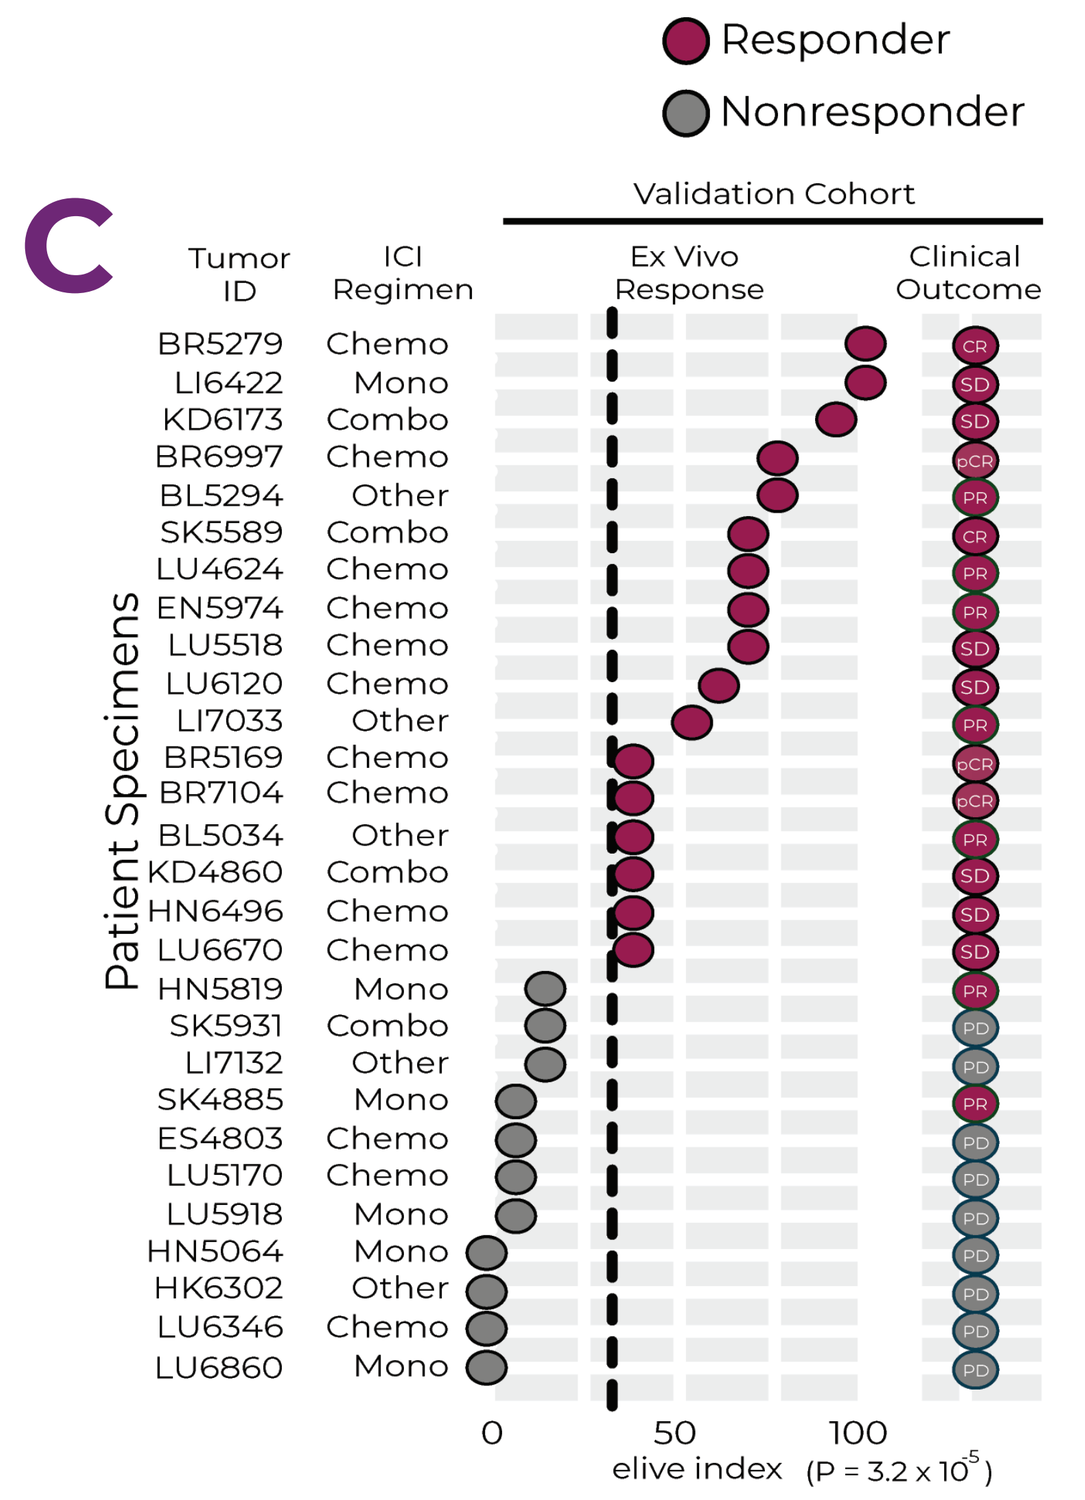
Responder
Nonresponder
Validation Cohort
Tumor ID
ICI Regimen
Ex Vivo Response
Clinical Outcome
Patient Specimens
BR5279 Chemo
LI6422 Mono
KD6173 Combo
BR6997 Chemo
BL5294 Other
SK5589 Combo
LU4624 Chemo
EN5974 Chemo
LU5518 Chemo
LU6120 Chemo
LI7033 Other
BR5169 Chemo
BR7104 Chemo
BL5034 Other
KD4860 Combo
HN6496 Chemo
LU6670 Chemo
HN5819 Mono
SK5931 Combo
LI7132 Other
SK4885 Mono
ES4803 Chemo
LU5170 Chemo
LU5918 Mono
HN5064 Mono
HK6302 Other
LU6346 Chemo
LU6860 Mono
elive index (P = 3.2 x 10⁵)

D

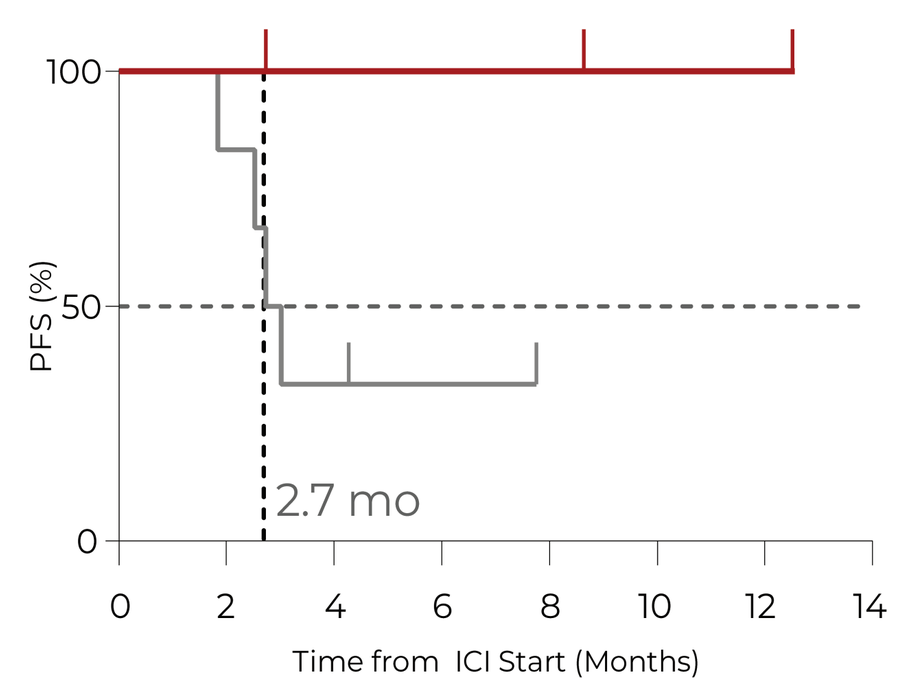
PFS (%)
2.7 mo
Time from ICI Start (Months)

E

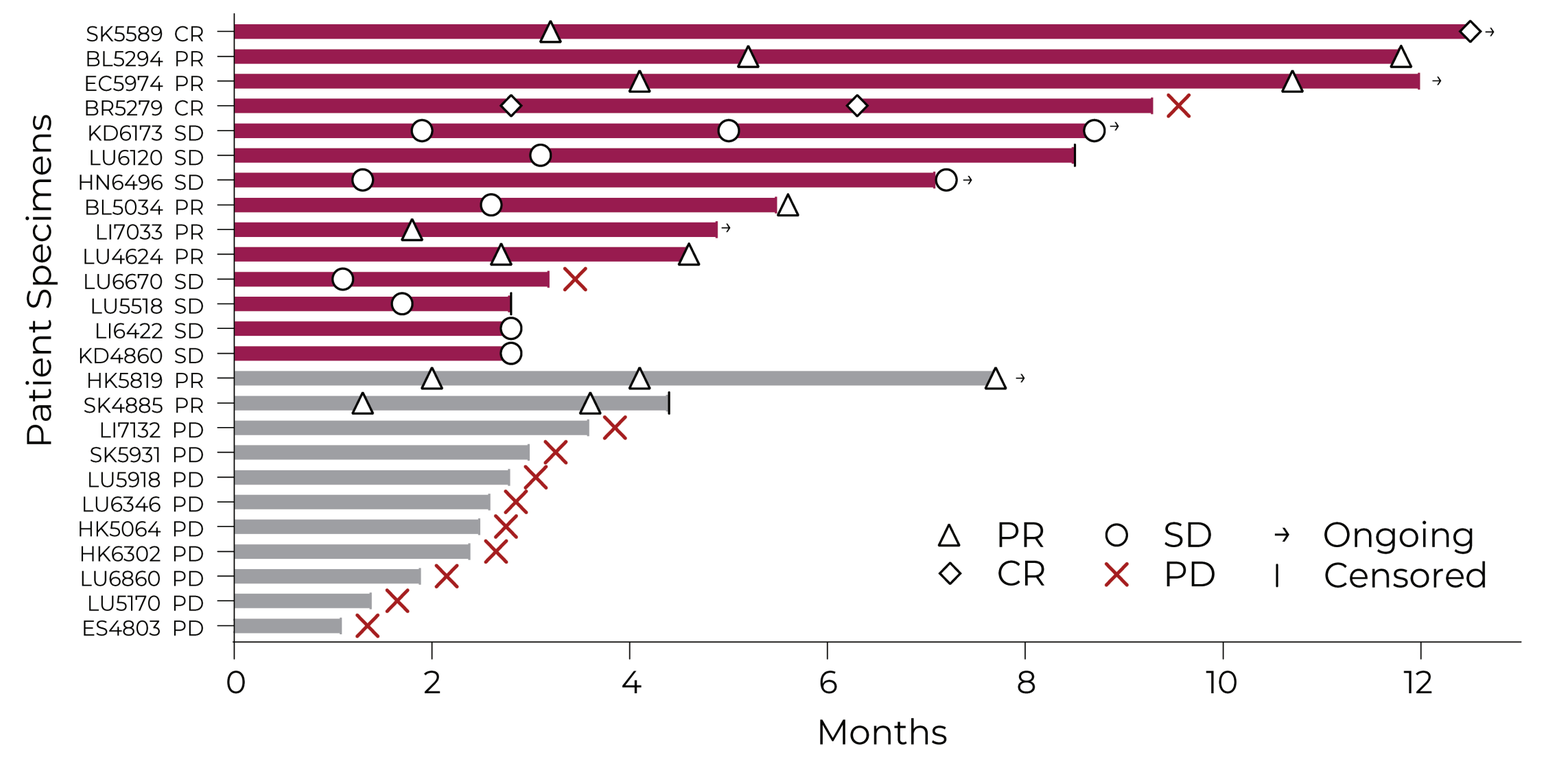
Patient Specimens
PR
CR
SD
PD
Ongoing
Censored
Months

A. Analysis of cytokine profiling data shown in Figure 1C identified the 8 most discriminative cytokines between the response and nonresponse clusters, which were used to develop a weighted classifier (elive index) that predicts sample classification within the response or nonresponse cluster. **B.** Area under the receiver operating characteristic of the elive index performance in predicting sample classification (response or non-response). **C.** elive index and associated clinical data from a validation cohort of 28 patients demonstrates that the elive index predicts clinical outcomes in patients receiving matched ICI treatment. **D.** Progression free survival (PFS) from time of ICI treatment initiation for elive index responders compared to non-responders in patients receiving pure ICI treatment. **E.** Swimmer plot comparing elive responders with non-responders, excluding patients in the neoadjuvant setting (PR = partial response, CR = complete response, SD = stable disease, PD = progressive disease).

3 Development of cytotoxicity endpoint broadens platform applications

A

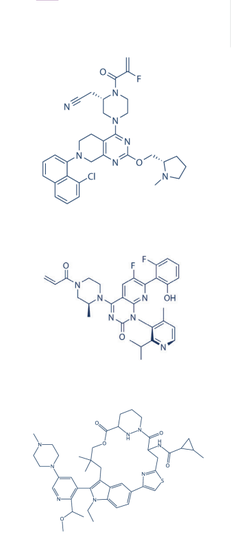

Drug	Mechanism of action	RAS state targeted	Mutations targeted
Adagrasib	Covalent	RAS(OFF)	G12C
Sotorasib	Covalent	RAS(OFF)	G12C
Daraxonrasib	Tri-complex	RAS(ON)	G12X, G13X, Q61X

B

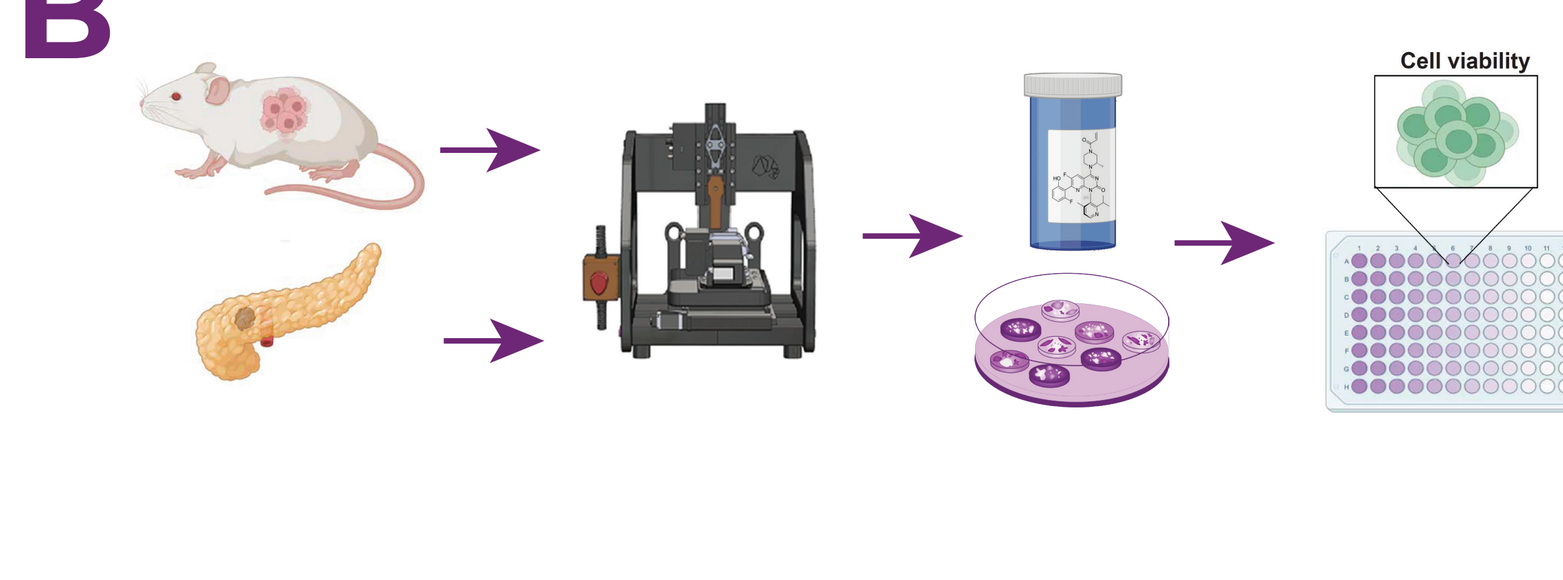
Cell viability

C

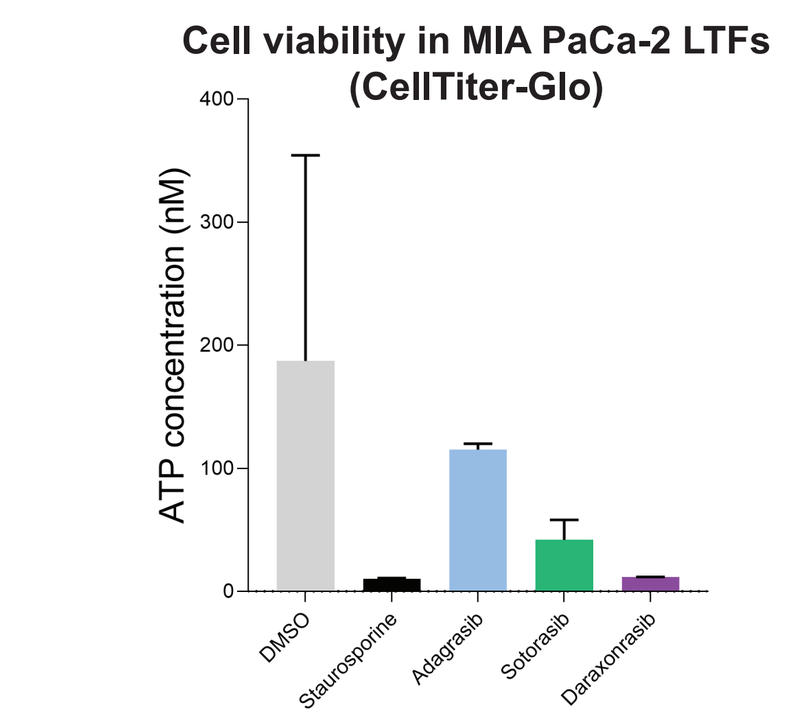
Cell viability in MIA PaCa-2 LTFs (CellTiter-Glo)
ATP concentration (nM)
DMSO
Staurosporine
Adagrasib
Sotorasib
Daraxonrasib

D

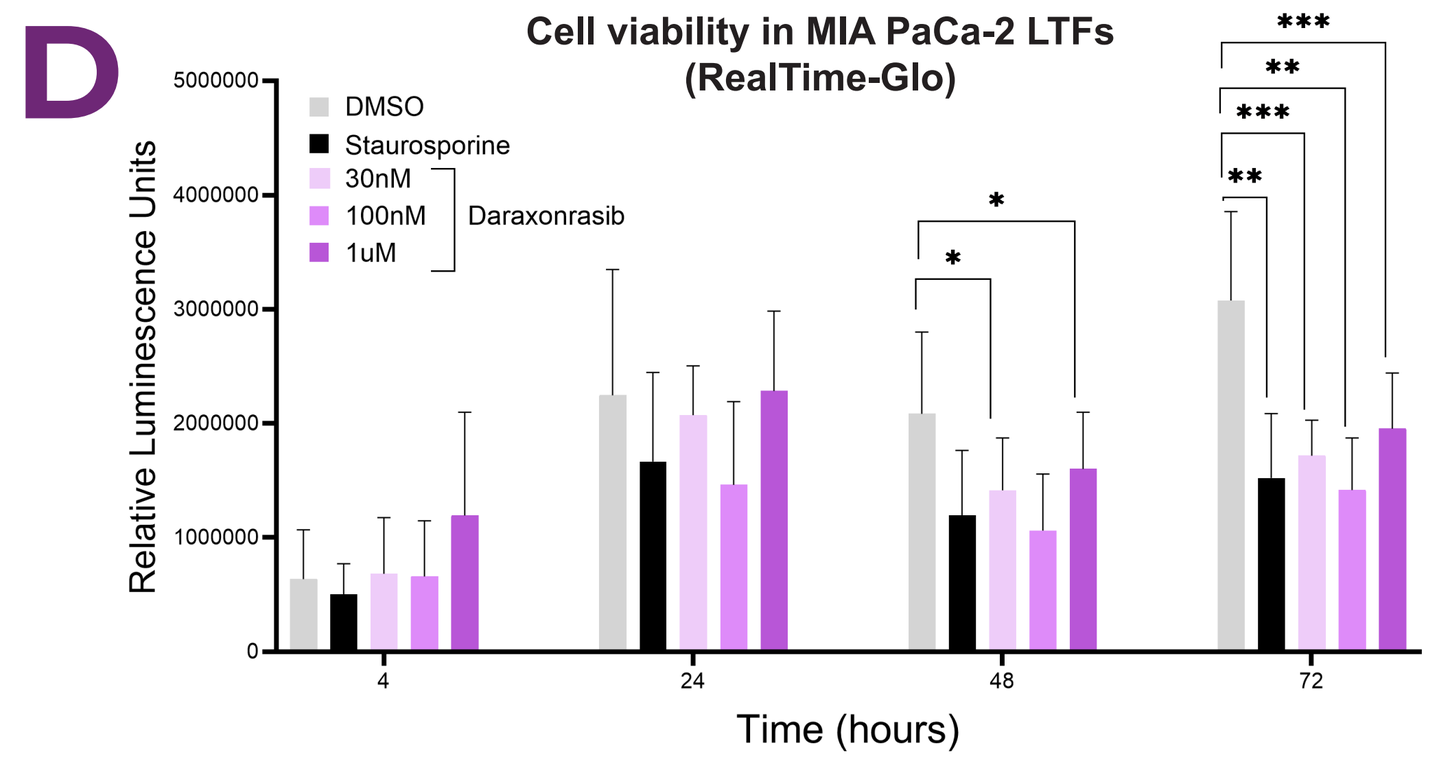
Cell viability in MIA PaCa-2 LTFs (RealTime-Glo)
Relative Luminescence Units
DMSO
Staurosporine
30nM
100nM
1uM
Daraxonrasib
Time (hours)

E

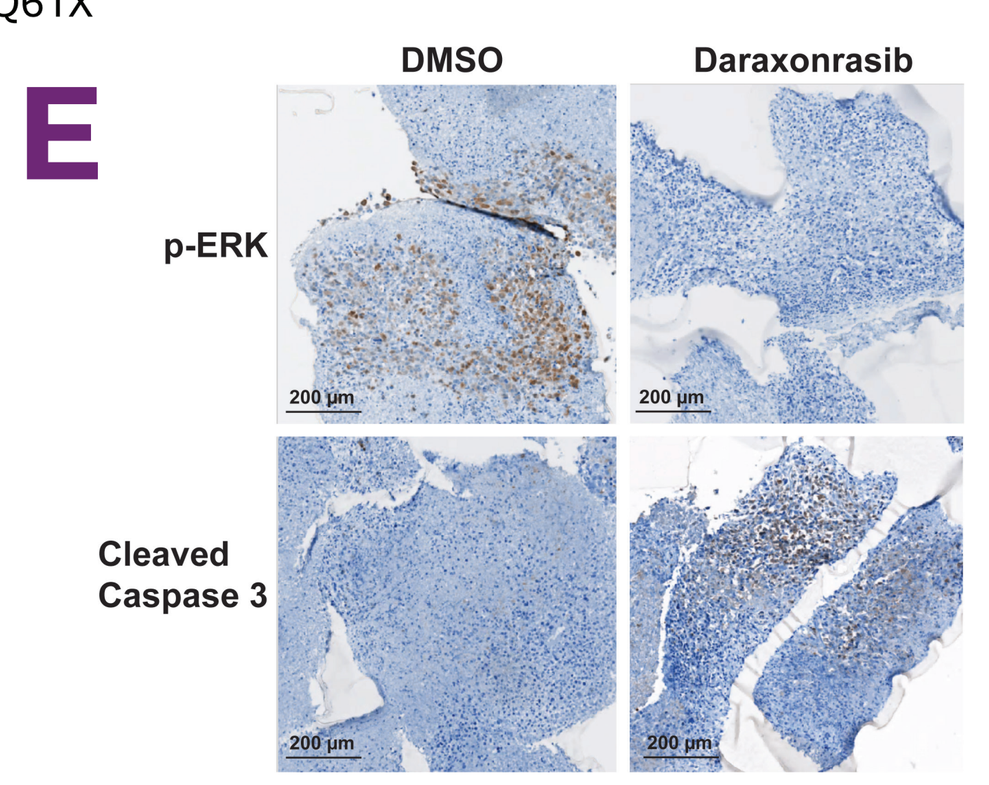
DMSO
Daraxonrasib
p-ERK
Cleaved Caspase 3
200 µm

F

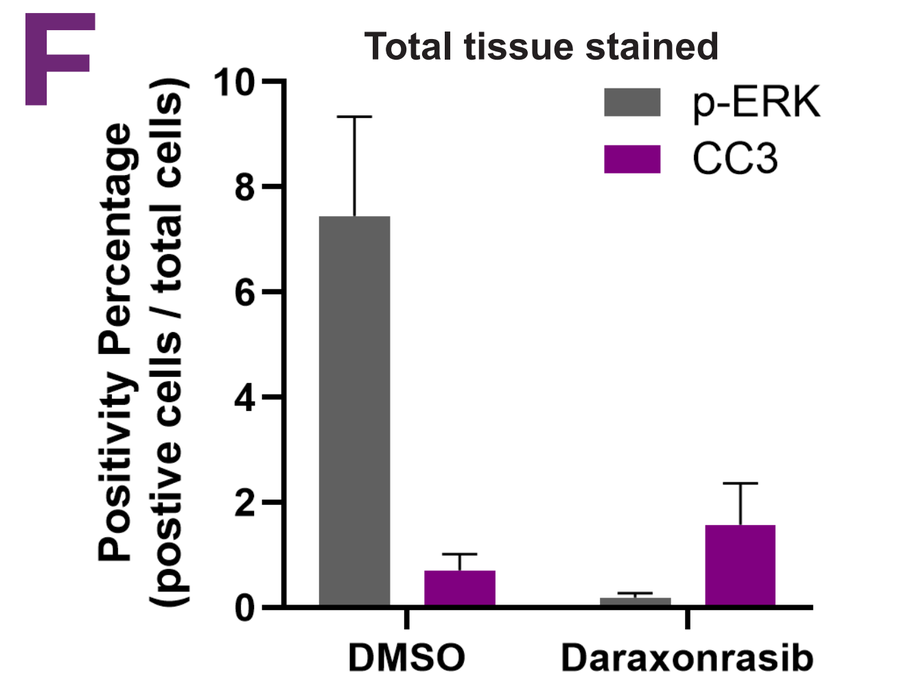
Total tissue stained
Positivity Percentage (postive cells / total cells)
p-ERK
CC3
DMSO
Daraxonrasib

G

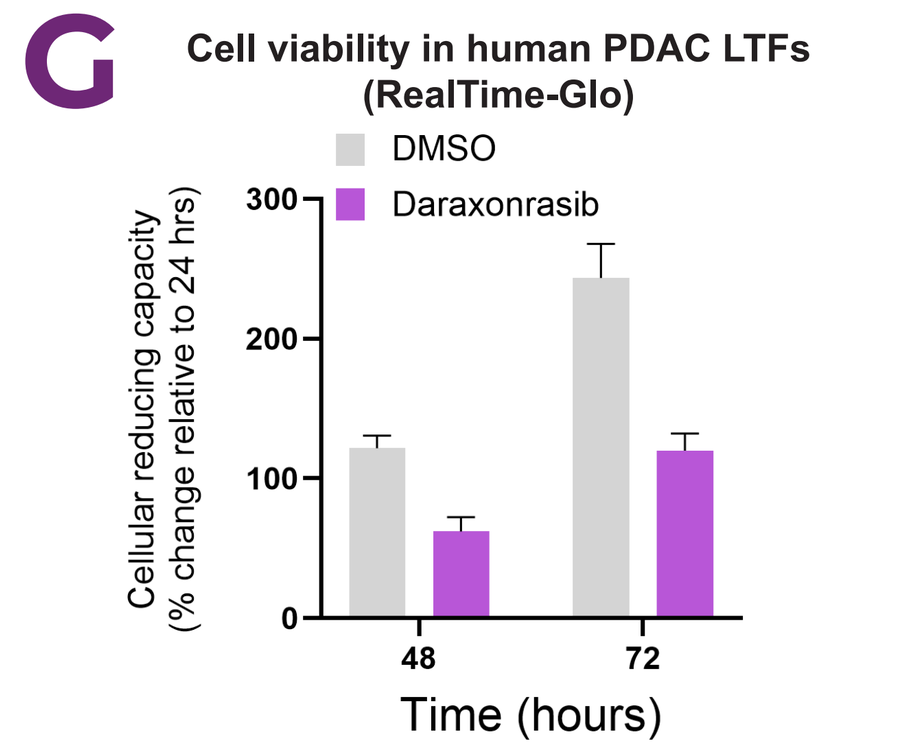
Cell viability in human PDAC LTFs (RealTime-Glo)
Cellular reducing capacity (% change relative to 24 hrs)
DMSO
Daraxonrasib
Time (hours)

A. Three KRAS inhibitors were used to characterize the cytotoxicity endpoint on the platform. **B.** Experimental setup: MIA PaCa-2 cells were implanted in immunocompromised mice to establish cell line–derived xenografts for use in the elive platform cytotoxicity assay. For human LTFs, a pancreatic resection specimen was used. Tumors were cut into LTFs using an automated cutting instrument designed for resections. LTFs were treated with the indicated KRAS inhibitor and downstream biochemical assays were used to assess treatment-induced effects on cell viability. **C-D.** Cell viability of MIA PaCa-2 LTFs treated with KRAS inhibitors (for C: all 1µM). **E.** Representative IHC images of p-ERK and cleaved caspase 3 (CC3) for MIA PaCa-2 LTFs treated with DMSO and 100nM daraxonrasib. **F.** Percent of cells positive for p-ERK and CC3 in MIA PaCa-2 LTFs for all replicates. **G.** Proof-of-concept experiment in a human pancreatic ductal adenocarcinoma (PDAC) specimen shows decreased cell viability in LTFs treated with 10 µM daraxonrasib compared with DMSO control.

References: **1.** Chen PL, et al. Cancer Discov. 2016;6(8):827-837. **2.** Pilco-Janeta D, De la Cruz-Puebla M. Anticancer Drugs. 2026;37(3):238-244. **3.** Voabil P, et al. Nat Med. 2021;27(7):1250-1261. **4.** Ramasubramanian TS, et al. J Transl Med. 2026;24(1):18. doi:10.1186/s12967-025-07378-2. **5.** Braun D, et al. in press, Cell Reports Medicine; medRxiv doi:10.64898/2026.06.05.26354635.